OR641 is a novel dual antagonist antibody that targets LILRB1 and LILRB2 inhibitory receptors and promotes a Th1-like immune response

Meghan Zuck, Kevin Green, Tatyana Pisarenko, Myriam Bouchlaka, Huyen Dinh, Francisco Zapata, Elsa Hay, Gajendra S. Naika, Jacob Heit, Ray Fox, Darbie Whitman, Tom Graddis, Kamal D. Puri, Peter Probst
OncoResponse Inc., 1124 Columbia Street, Suite 300, Seattle, WA 98104, USA

Abstract # 519

Background: The immunosuppression of myeloid cells and lymphocytes within the tumor microenvironment (TME) limits efficacy of checkpoint inhibitors. LILRB1 (ILT2) and LILRB2 (ILT4) are inhibitory receptors on immune cells that interact with ligands including classical and nonclassical HLA Class I (e.g., HLA-A, HLA-G). LILRB1 and LILRB2 are expressed on myeloid cells, and LILRB1 is additionally expressed on subsets of B, NK, and T cells. Interaction of LILRB1 and LILRB2 receptors with their ligands promotes an immunosuppressive phenotype of myeloid cells and inhibits T and NK cell cytotoxic activity required for tumor cell death. The LILRB1 receptor on macrophages contributes to “don’t eat me” signals for cancer cells to evade phagocytosis by macrophages. Dual antagonism of LILRB1 and LILRB2 by a single antibody to restore both innate and adaptive immune responses is a promising strategy to enhance efficacy of checkpoint inhibitors. A dual LILRB1/2 antagonist antibody is currently being evaluated in clinical trials for cancer treatment. We have identified OR641 as a dual antagonist antibody that demonstrates superior activity in relieving LILRB1- and LILRB2-mediated immune suppression and enhances both innate and adaptive anti-tumor immunity.

Methods: OR641 is a humanized antibody derived from rabbit B cells immunized with LILRB2 protein. The antibody was evaluated for its activity in various biochemical and in vitro pharmacological assays using primary human macrophages, T cells and NK cells. The pharmacokinetic profile of OR641 was assessed in humanized FcRn mice.

Results: OR641 binds specifically to human LILRB1 and LILRB2 proteins and blocks their interactions with HLA class I ligands. OR641 demonstrated superior activity to other antibodies in pharmacological assays modeling LILRB1 and LILRB2-mediated immunosuppression in the TME. OR641 promoted a Th1-like innate immune response by enhancing IFN-γ production and decreasing IL-10 secretion by PBMCs stimulated with TLR ligands. Treatment with OR641 restored the ability of peripheral blood T cells and exhausted T cells to secrete IFN-γ in the presence of suppressive macrophages. OR641 enhanced macrophage phagocytosis of HLA-G+ tumor cells, and rescued the cytotoxic activity of NK cells from LILRB1 mediated immune suppression. The half-life of OR641 in humanized FcRn mice was 10 days.

Conclusions: OR641 is a unique dual anti-LILRB2/1 antagonist antibody that promotes a Th1-like immune response. OR641 modulates immunosuppressive myeloid cells through blockade of LILRB1 and LILRB2 and stimulates both innate and adaptive immune responses. These data provide a strong rationale for further development of OR641 as a treatment for solid tumor malignancies.

LILRB1 and LILRB2 dual antagonism drives anti-tumor activity

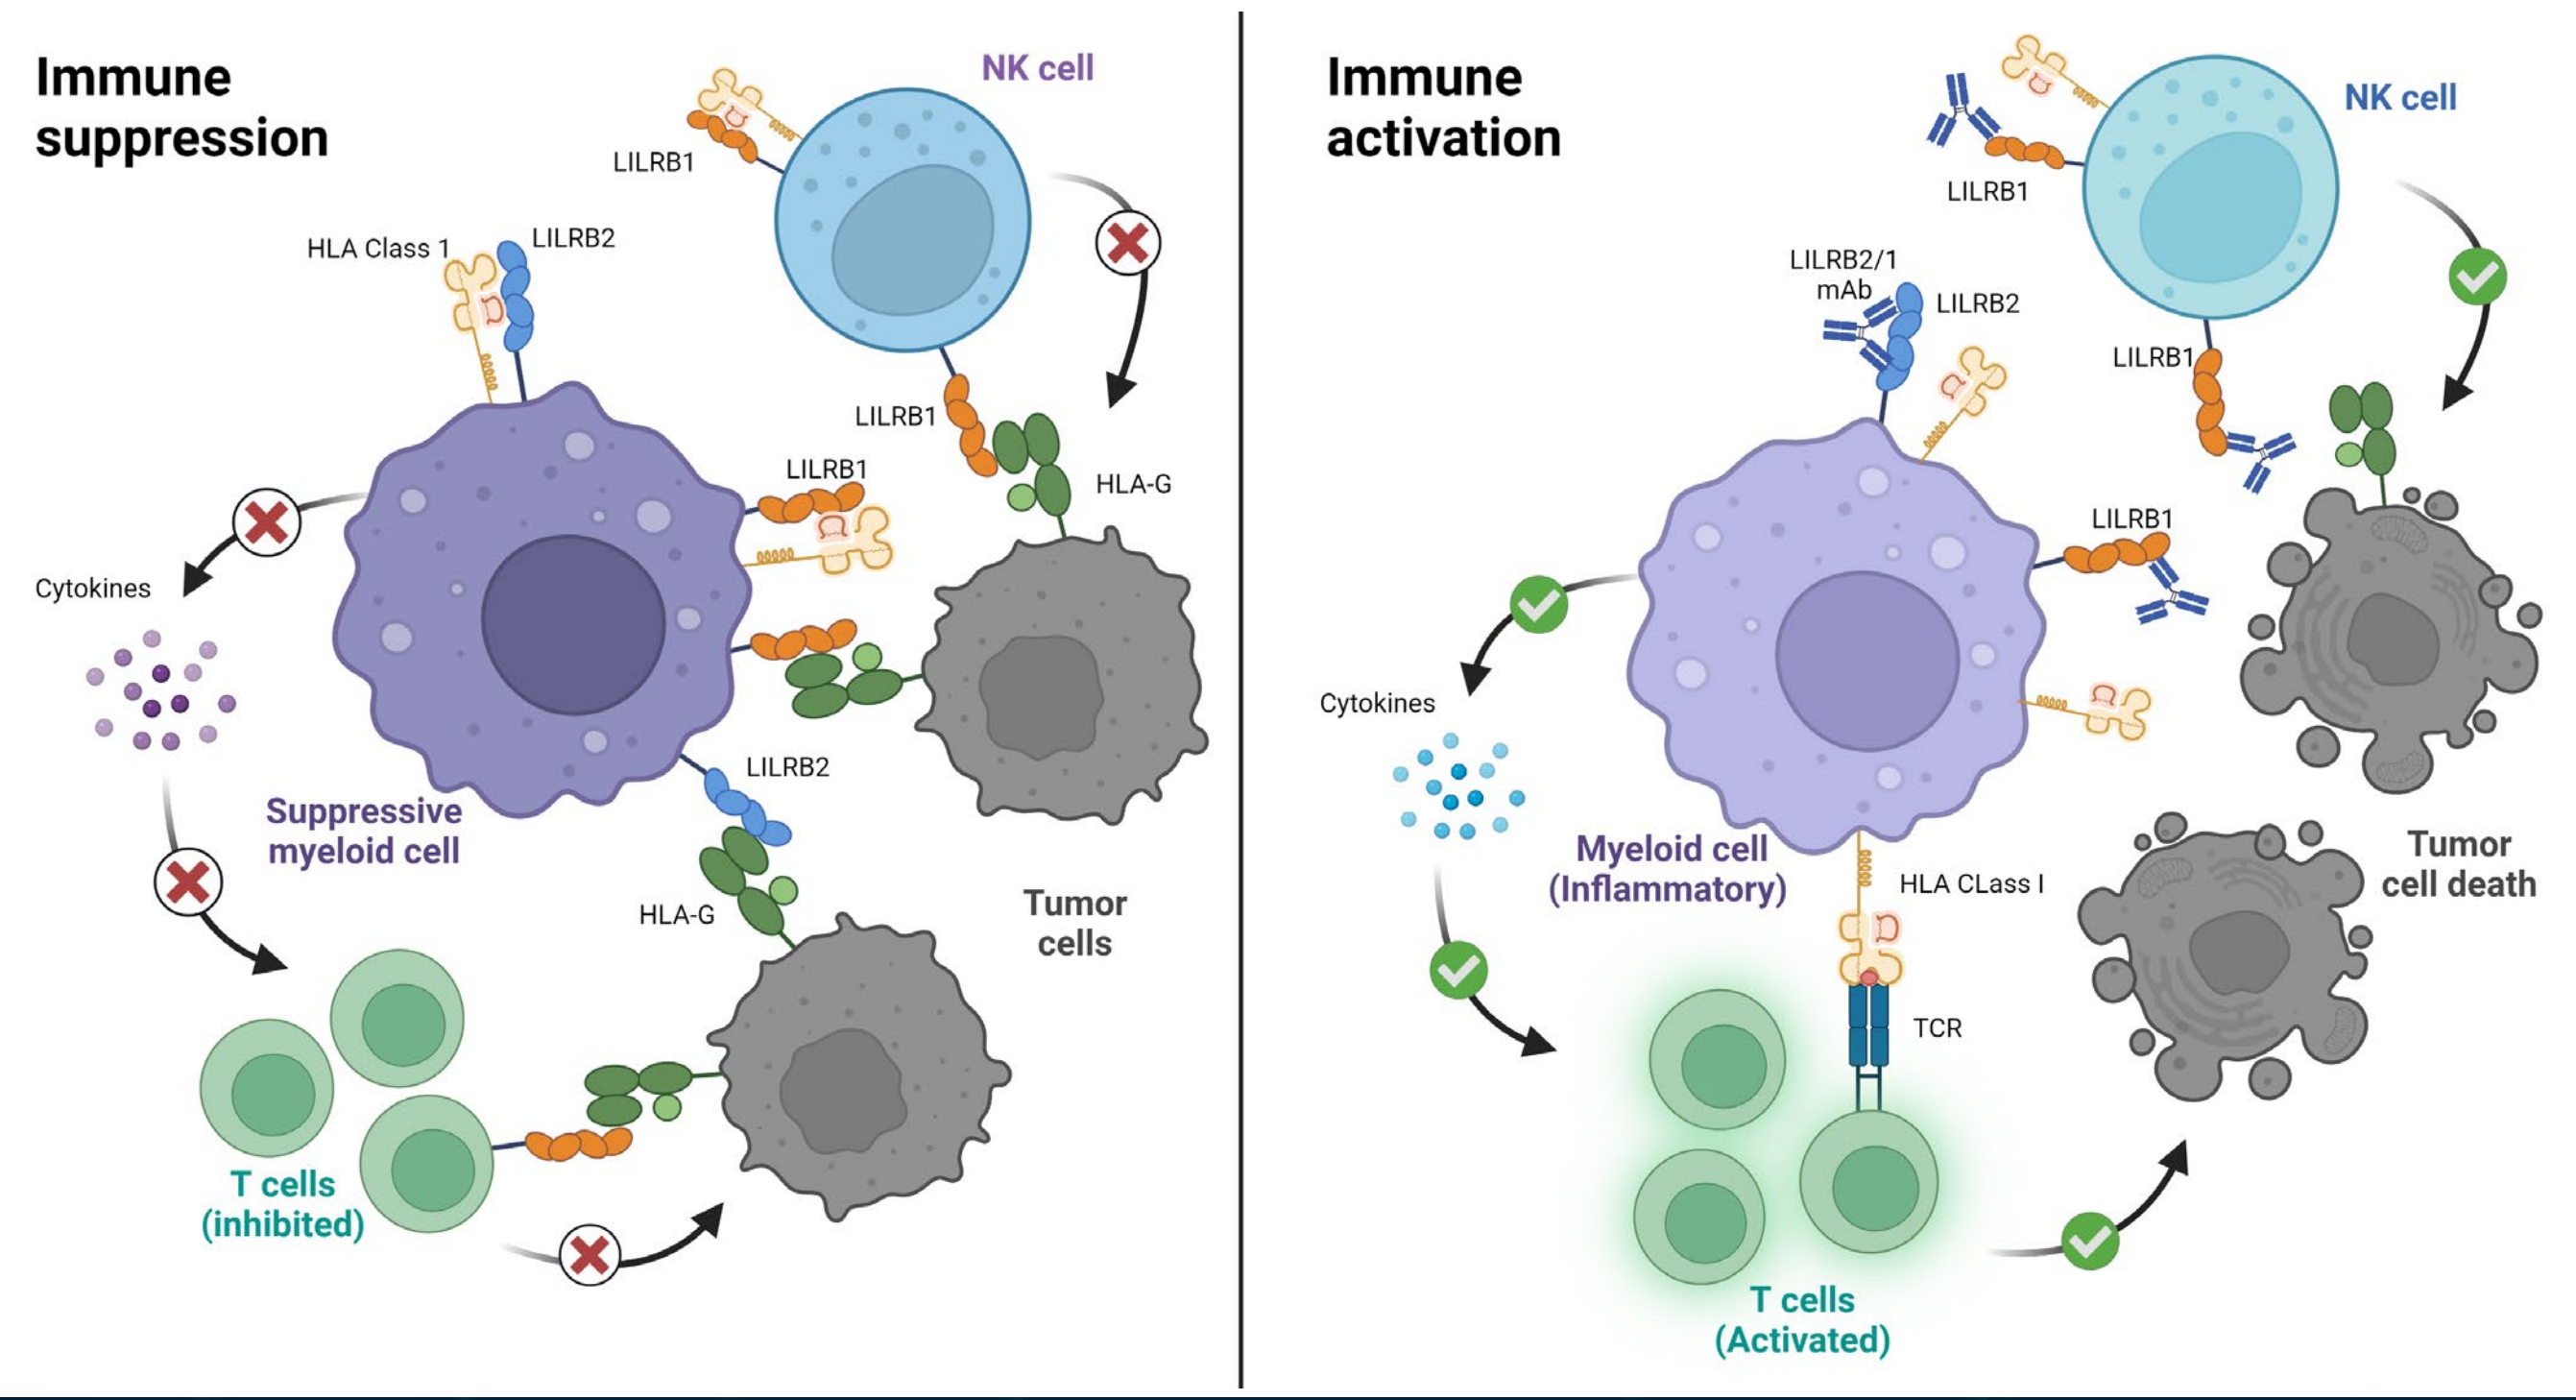
Immune suppression
NK cell
LILRB1
HLA Class 1
LILRB2
HLA-G
Cytokines
Suppressive myeloid cell
Tumor cells
T cells (inhibited)
Immune activation
LILRB2/1 mAb
Myeloid cell (Inflammatory)
HLA CLass I
TCR
Tumor cell death
T cells (Activated)

RESULTS

OR641 binds to LILRB1 and LILRB2 proteins and to myeloid cells

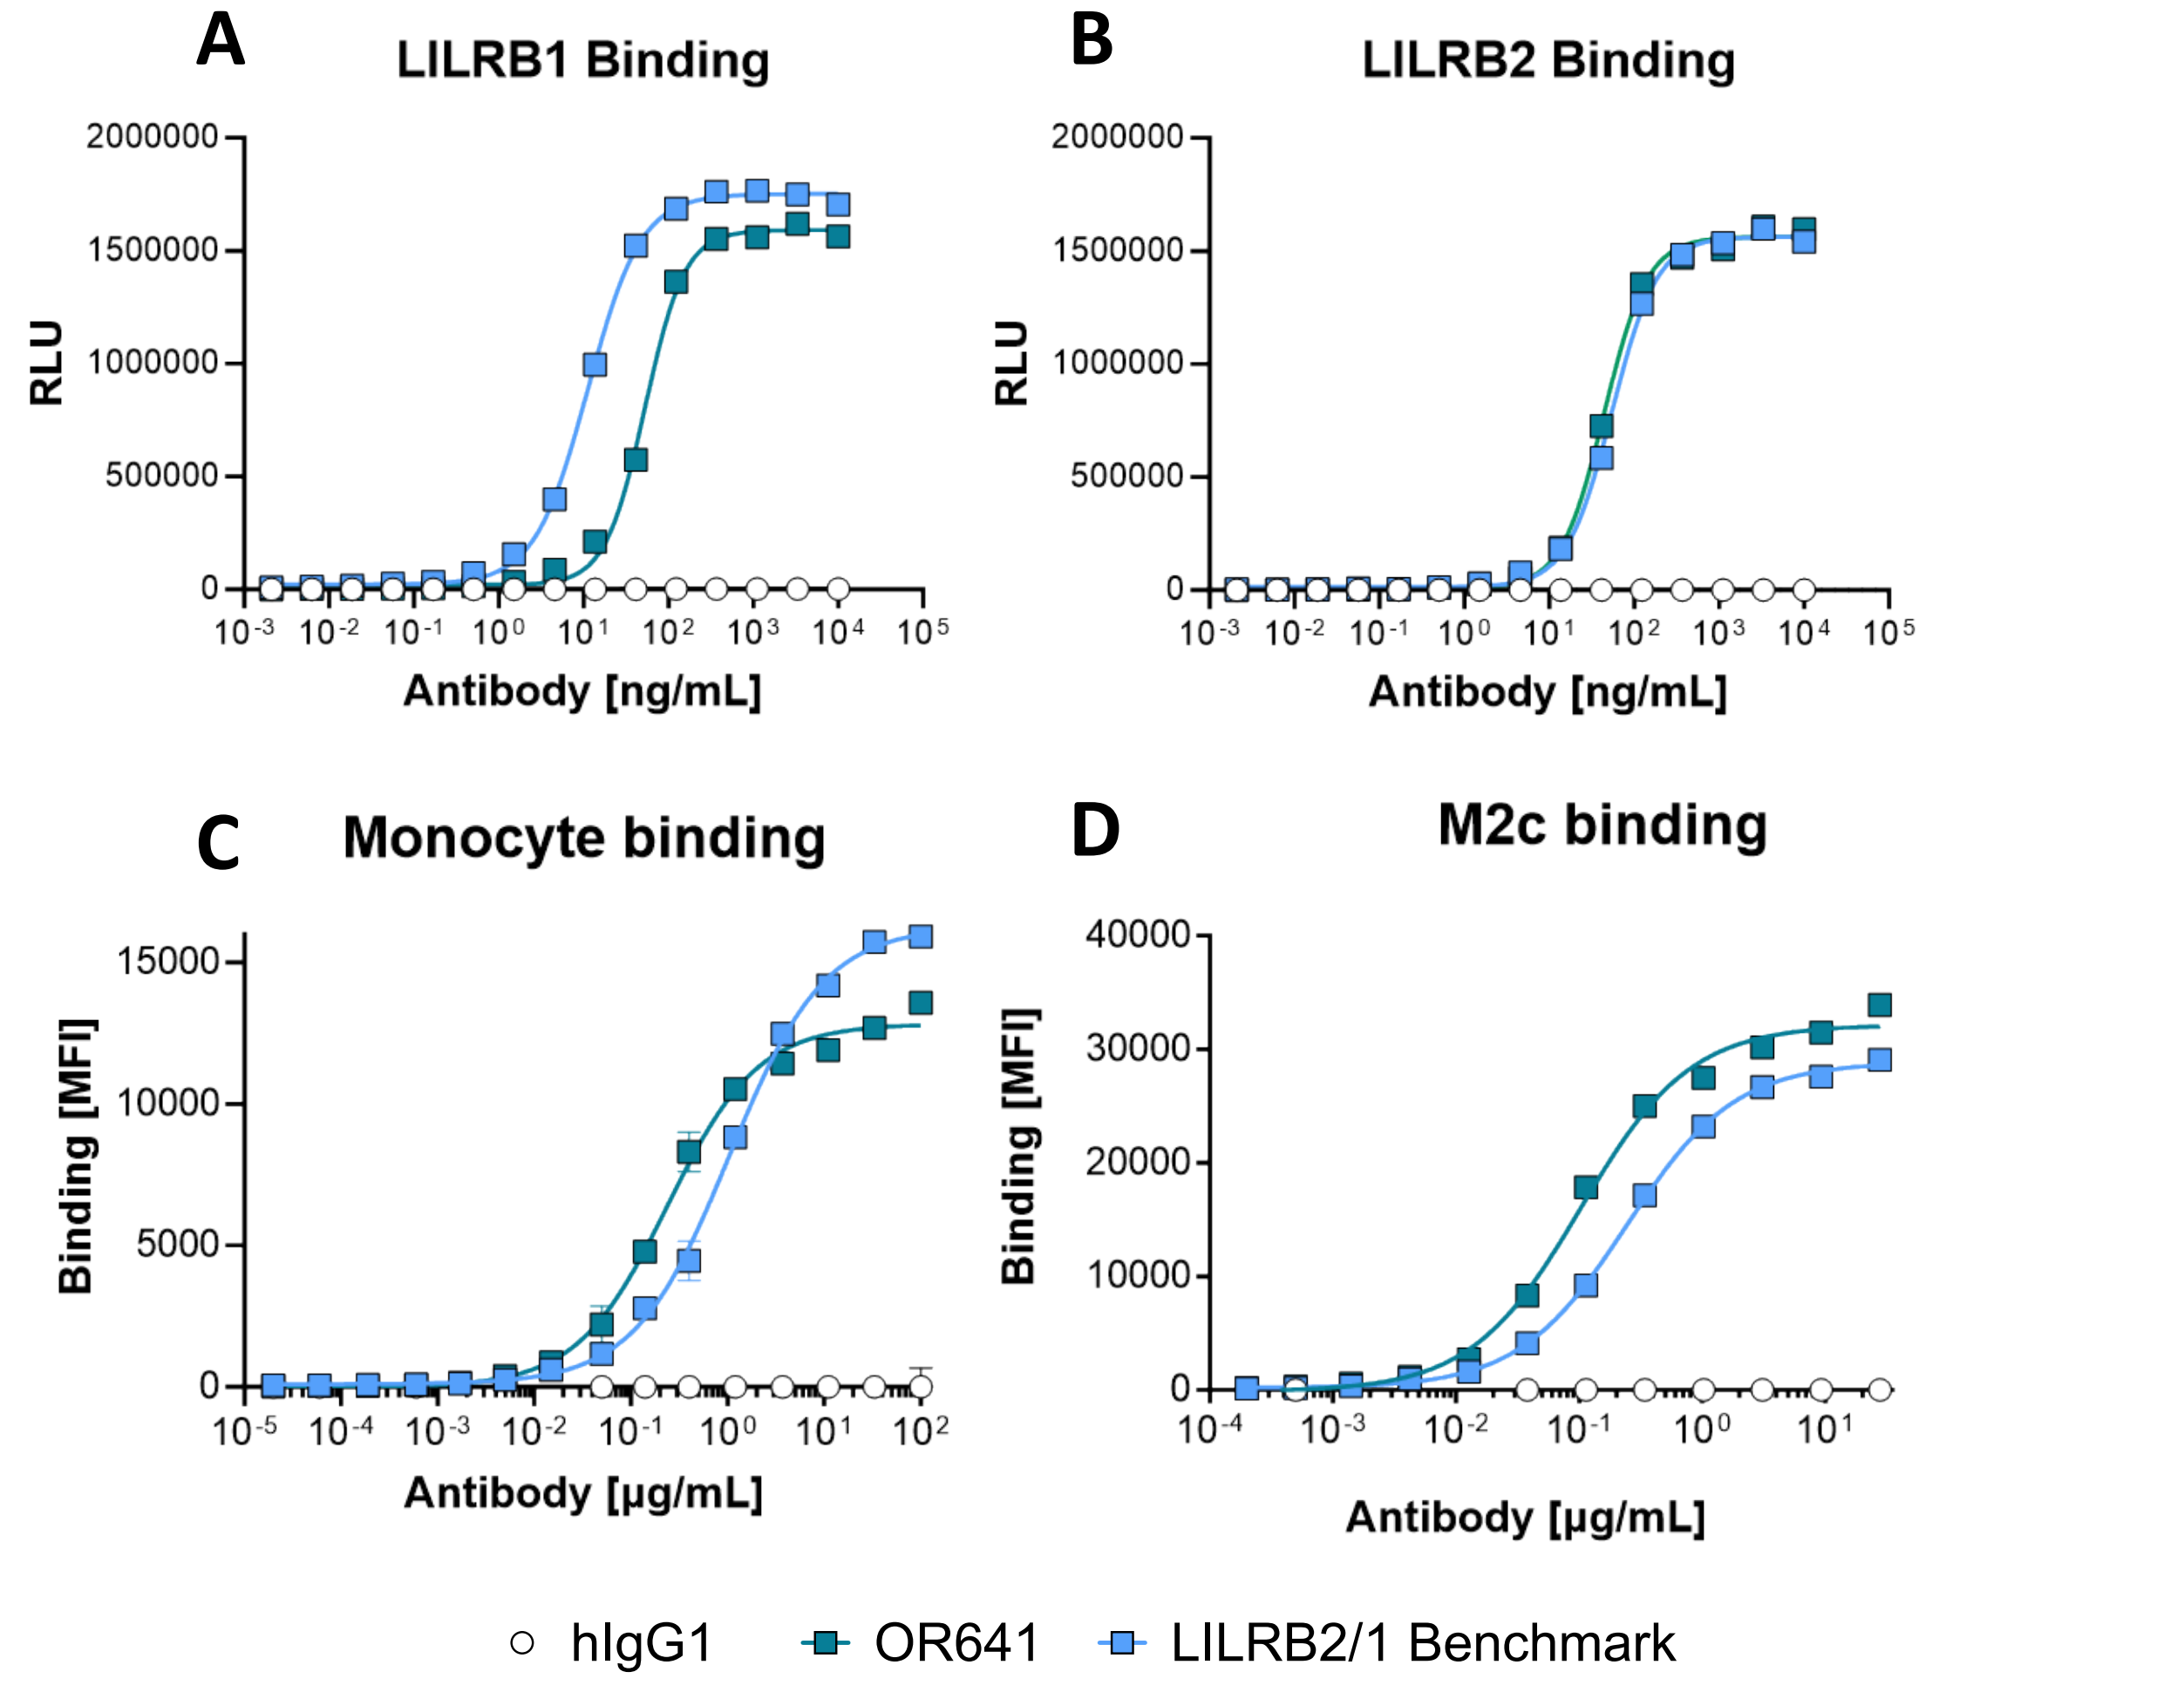
A
LILRB1 Binding
RLU
Antibody [ng/mL]
B
LILRB2 Binding
C
Monocyte binding
Binding [MFI]
Antibody [μg/mL]
D
M2c binding
hIgG1
OR641
LILRB2/1 Benchmark

Figure 2. OR641 binds to A) human LILRB1 and B) human LILRB2 by ELISA. OR641 binds to C) human primary monocytes and D) human M2c macrophages, measured via flow cytometry.

OR641 blocks binding of human LILRB1 and LILRB2 to HLA-G on 721.221 B-cell lymphoma cells

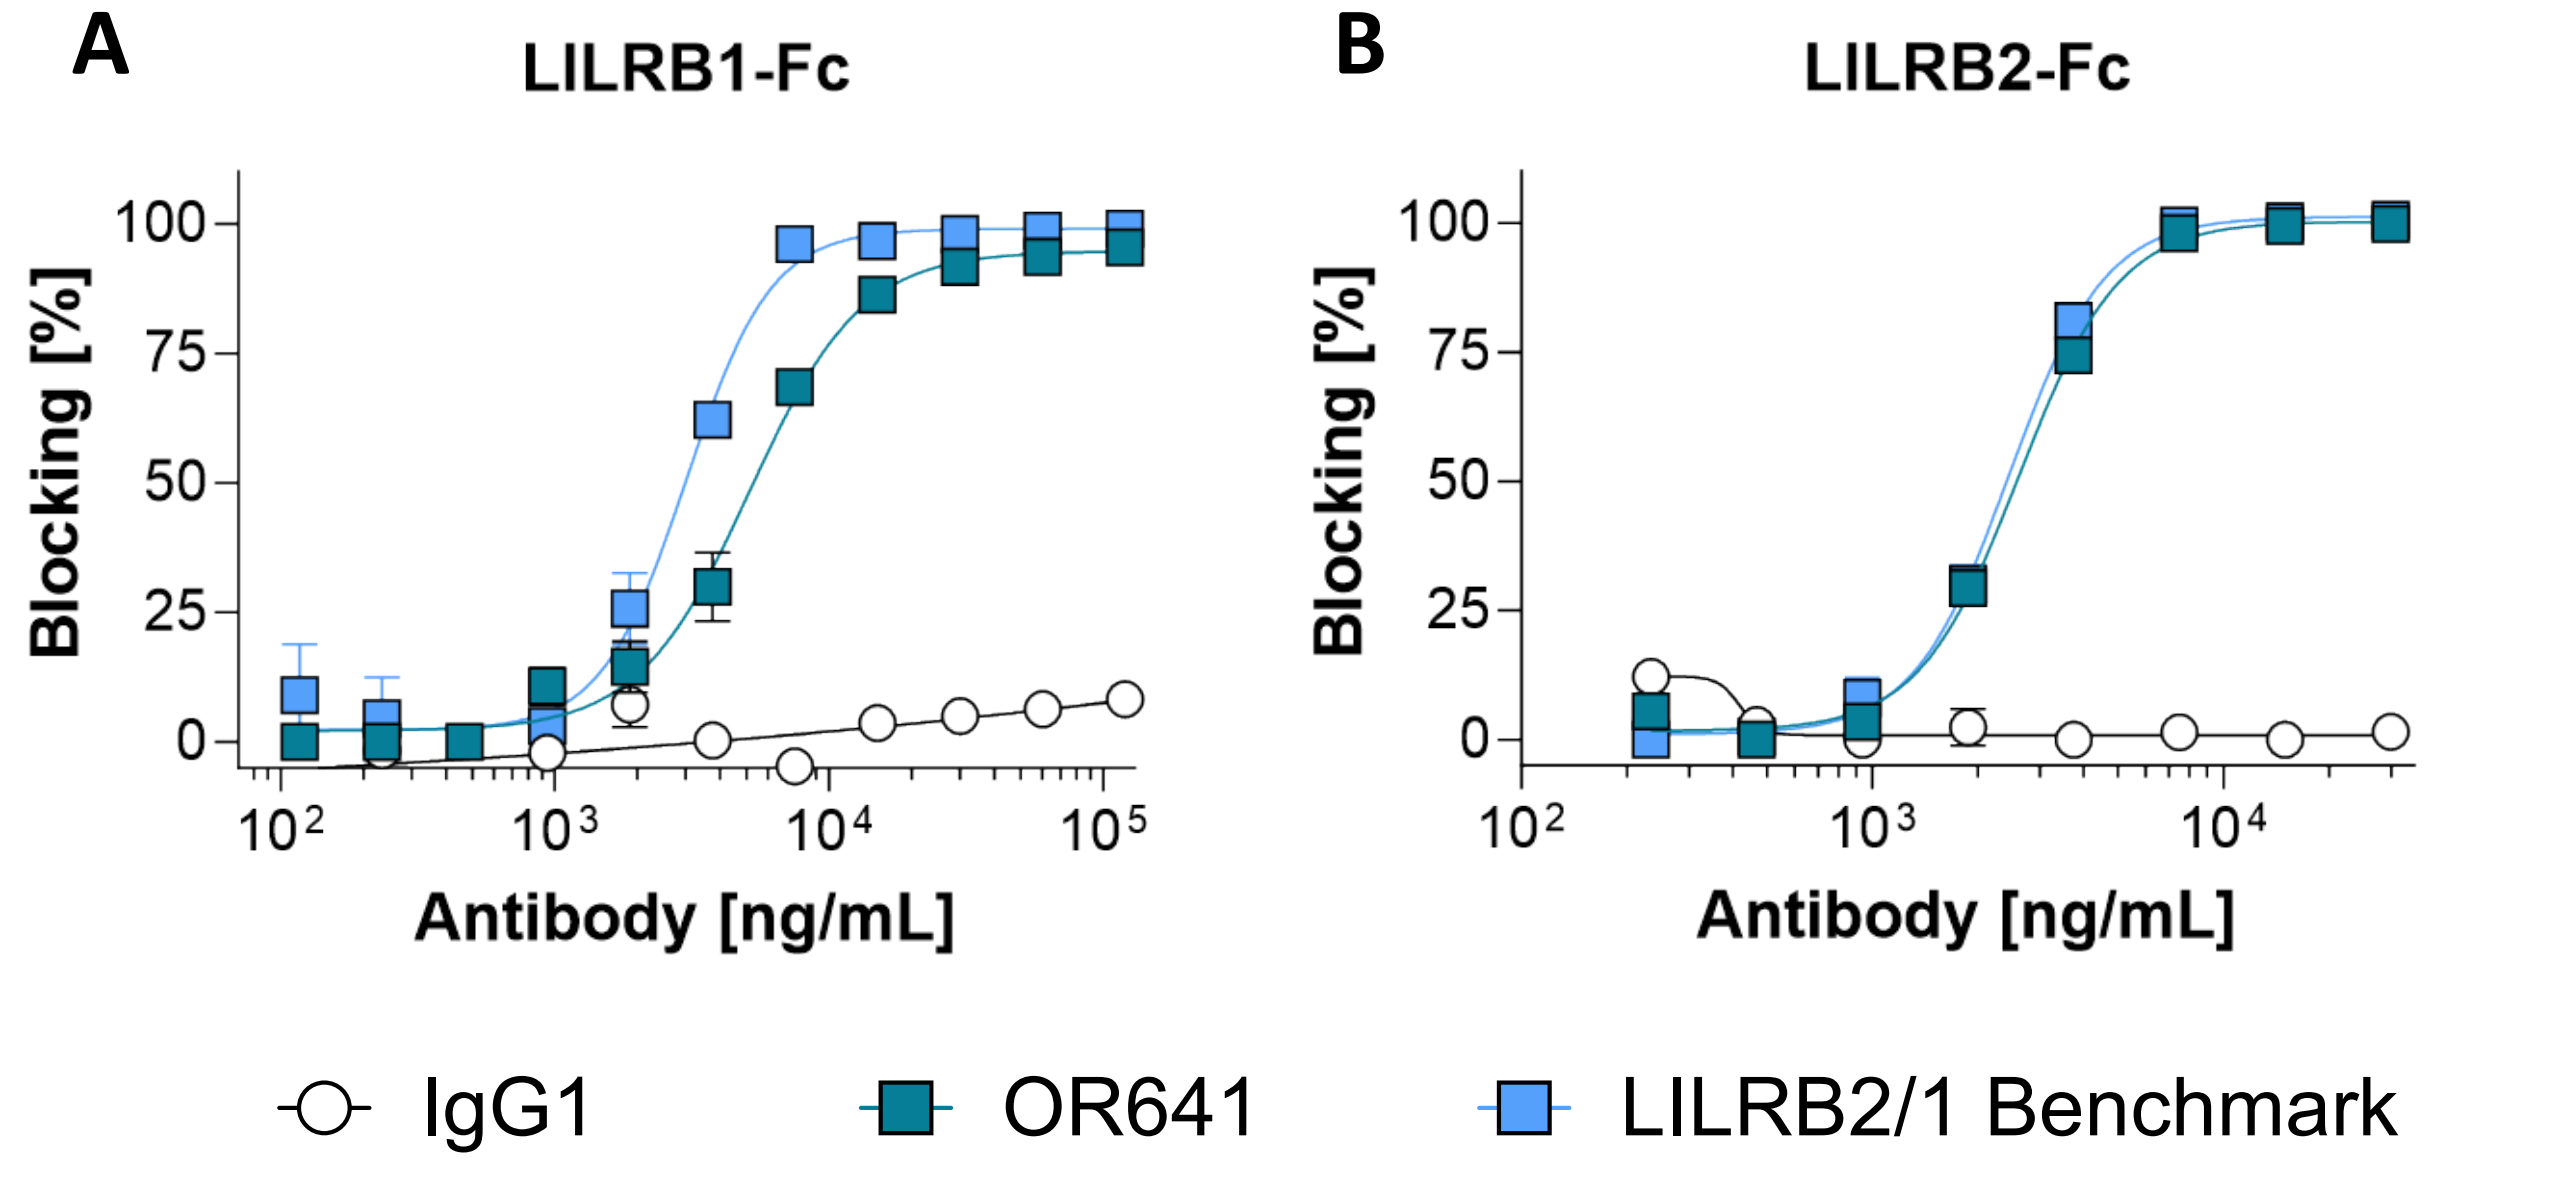
A
LILRB1-Fc
B
LILRB2-Fc
Blocking [%]
Antibody [ng/mL]
IgG1
OR641
LILRB2/1 Benchmark

Figure 3. OR641 blocks the interaction of A) human LILRB1-Fc and B) human LILRB2-Fc with HLA-G expressed on B-cell lymphoma 721.221 cells.

OR641 treatment relieves CD8⁺ T cells from M2c macrophage-mediated immune suppression

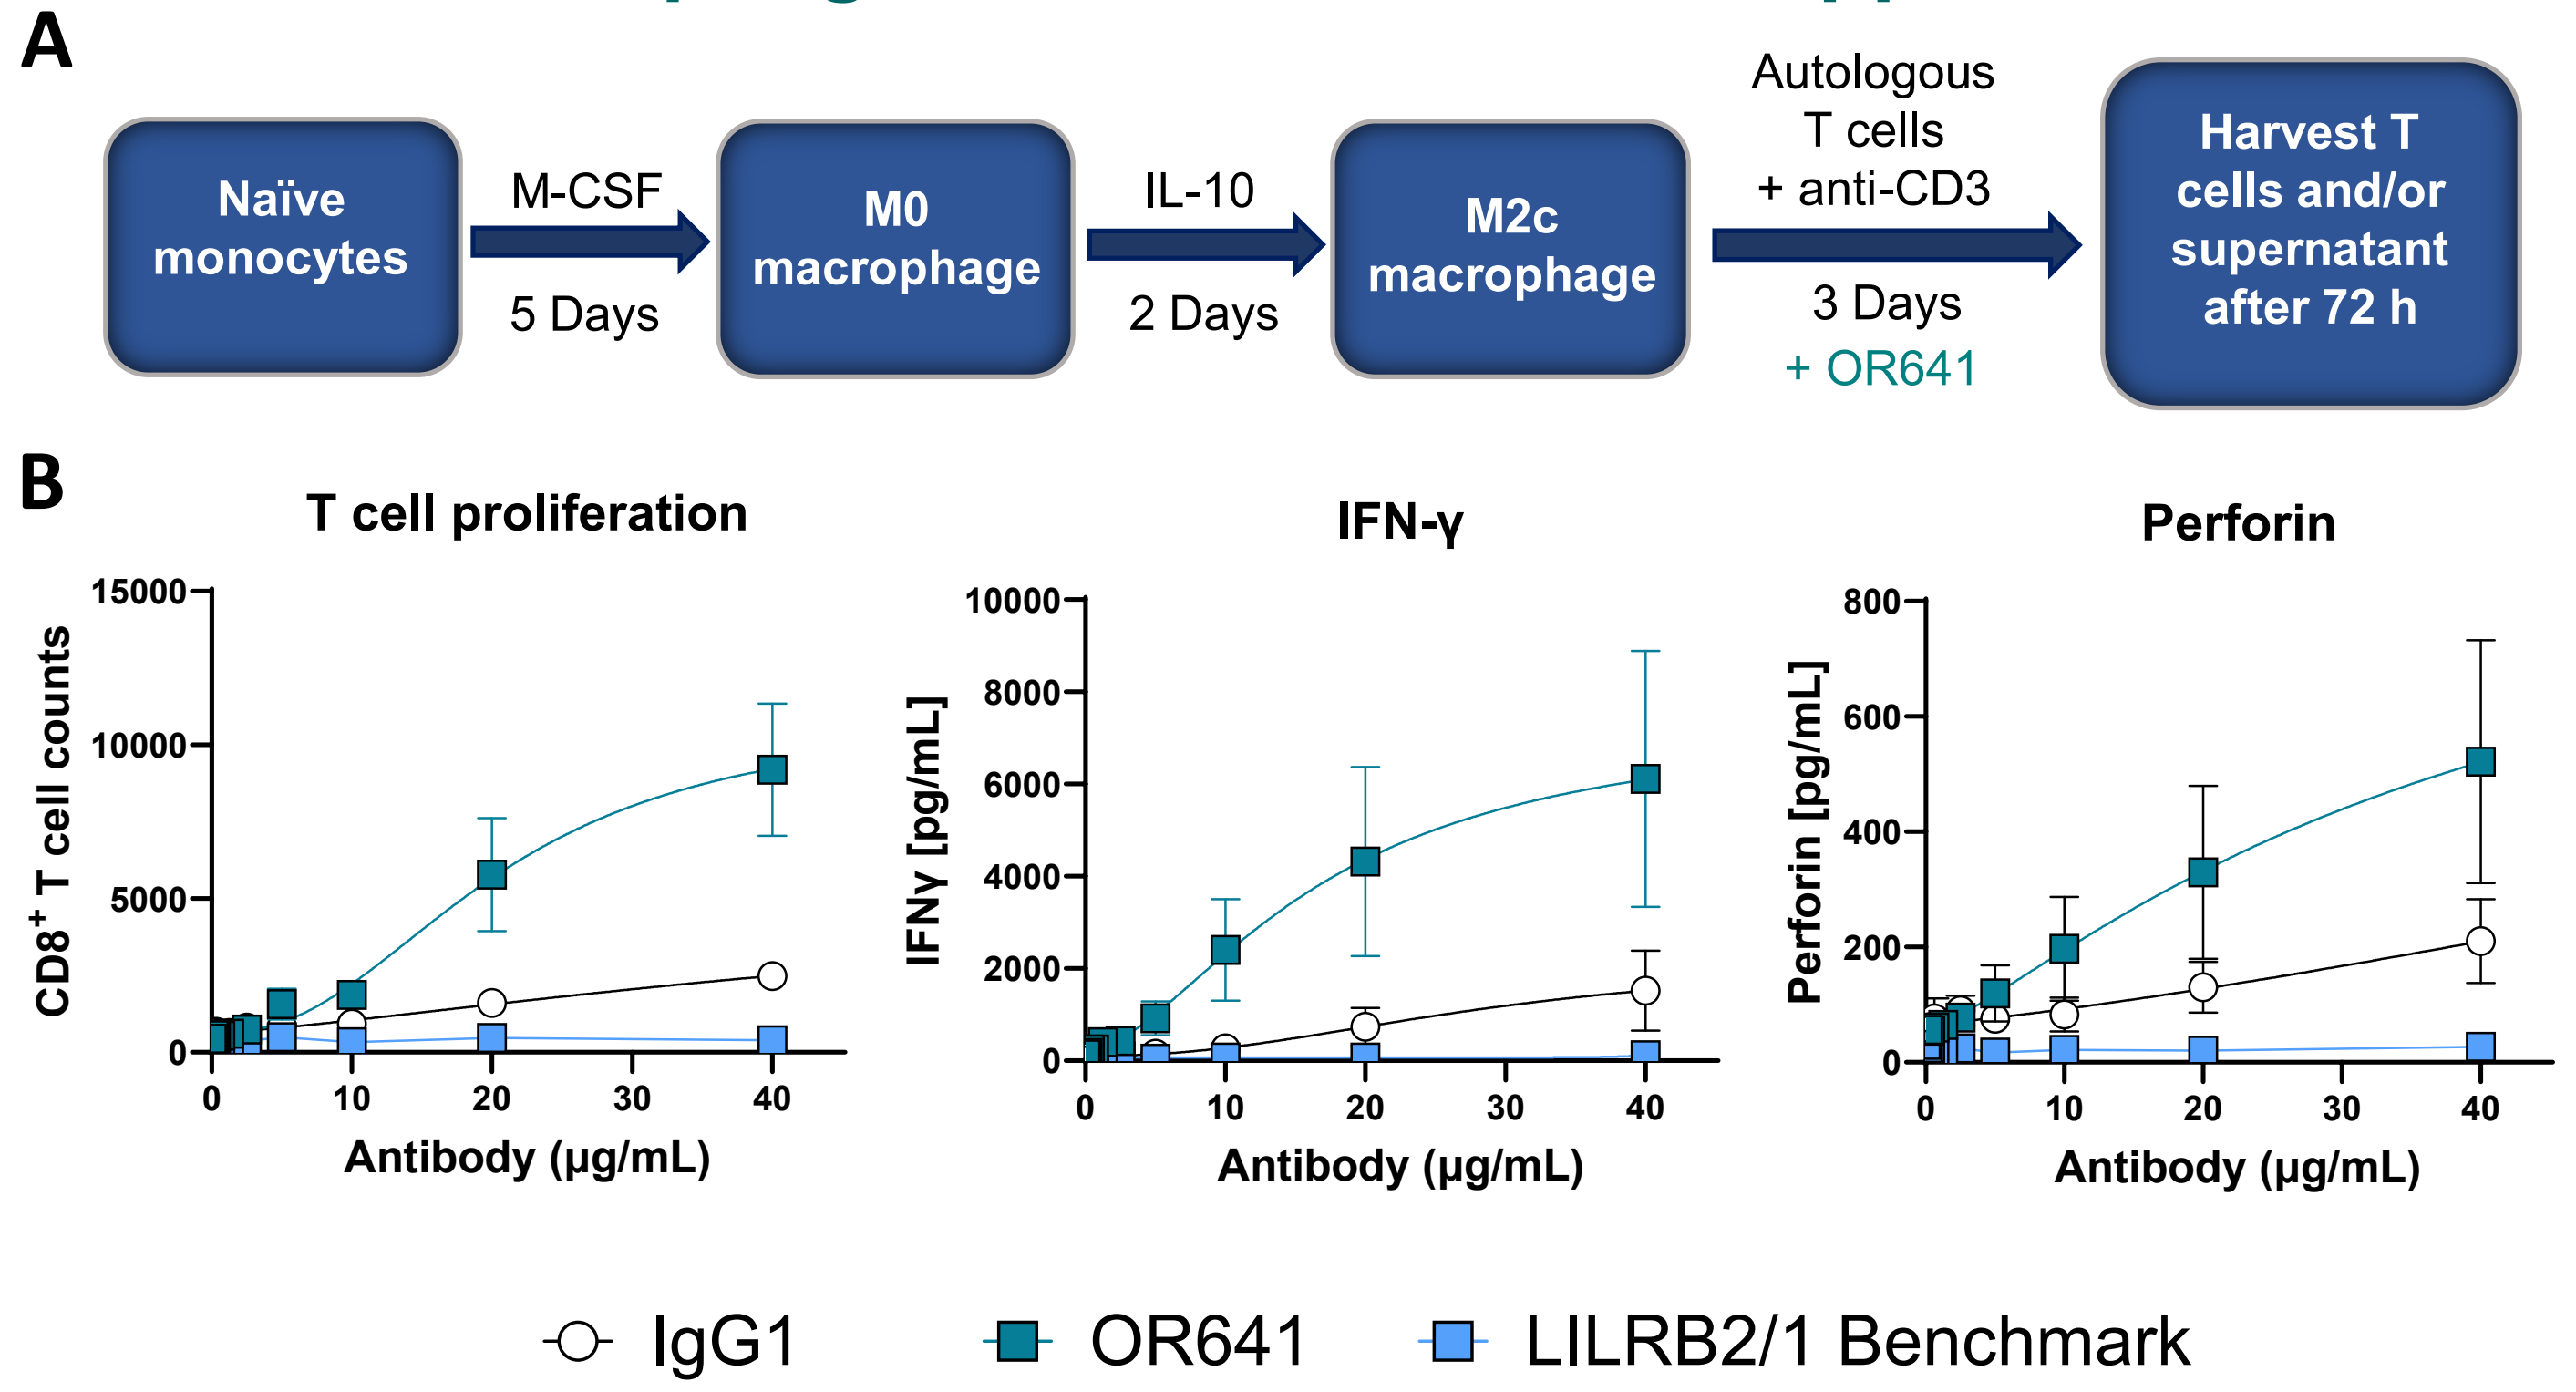
A
Naïve monocytes
M-CSF
5 Days
M0 macrophage
IL-10
2 Days
M2c macrophage
Autologous T cells + anti-CD3
3 Days
+ OR641
Harvest T cells and/or supernatant after 72 h
B
T cell proliferation
CD8⁺ T cell counts
IFN-γ
IFNγ [pg/mL]
Perforin
Perforin [pg/mL]
Antibody (μg/mL)
IgG1
OR641
LILRB2/1 Benchmark

Figure 4. A) M2c macrophage/CD8 T cell coculture assay procedure. B) Treatment with OR641 rescues T cells from M2c macrophage-mediated immune suppression.

OR641 treatment enhances IFN-γ secretion and decreases IL-10 secretion in human PBMCs stimulated with LPS

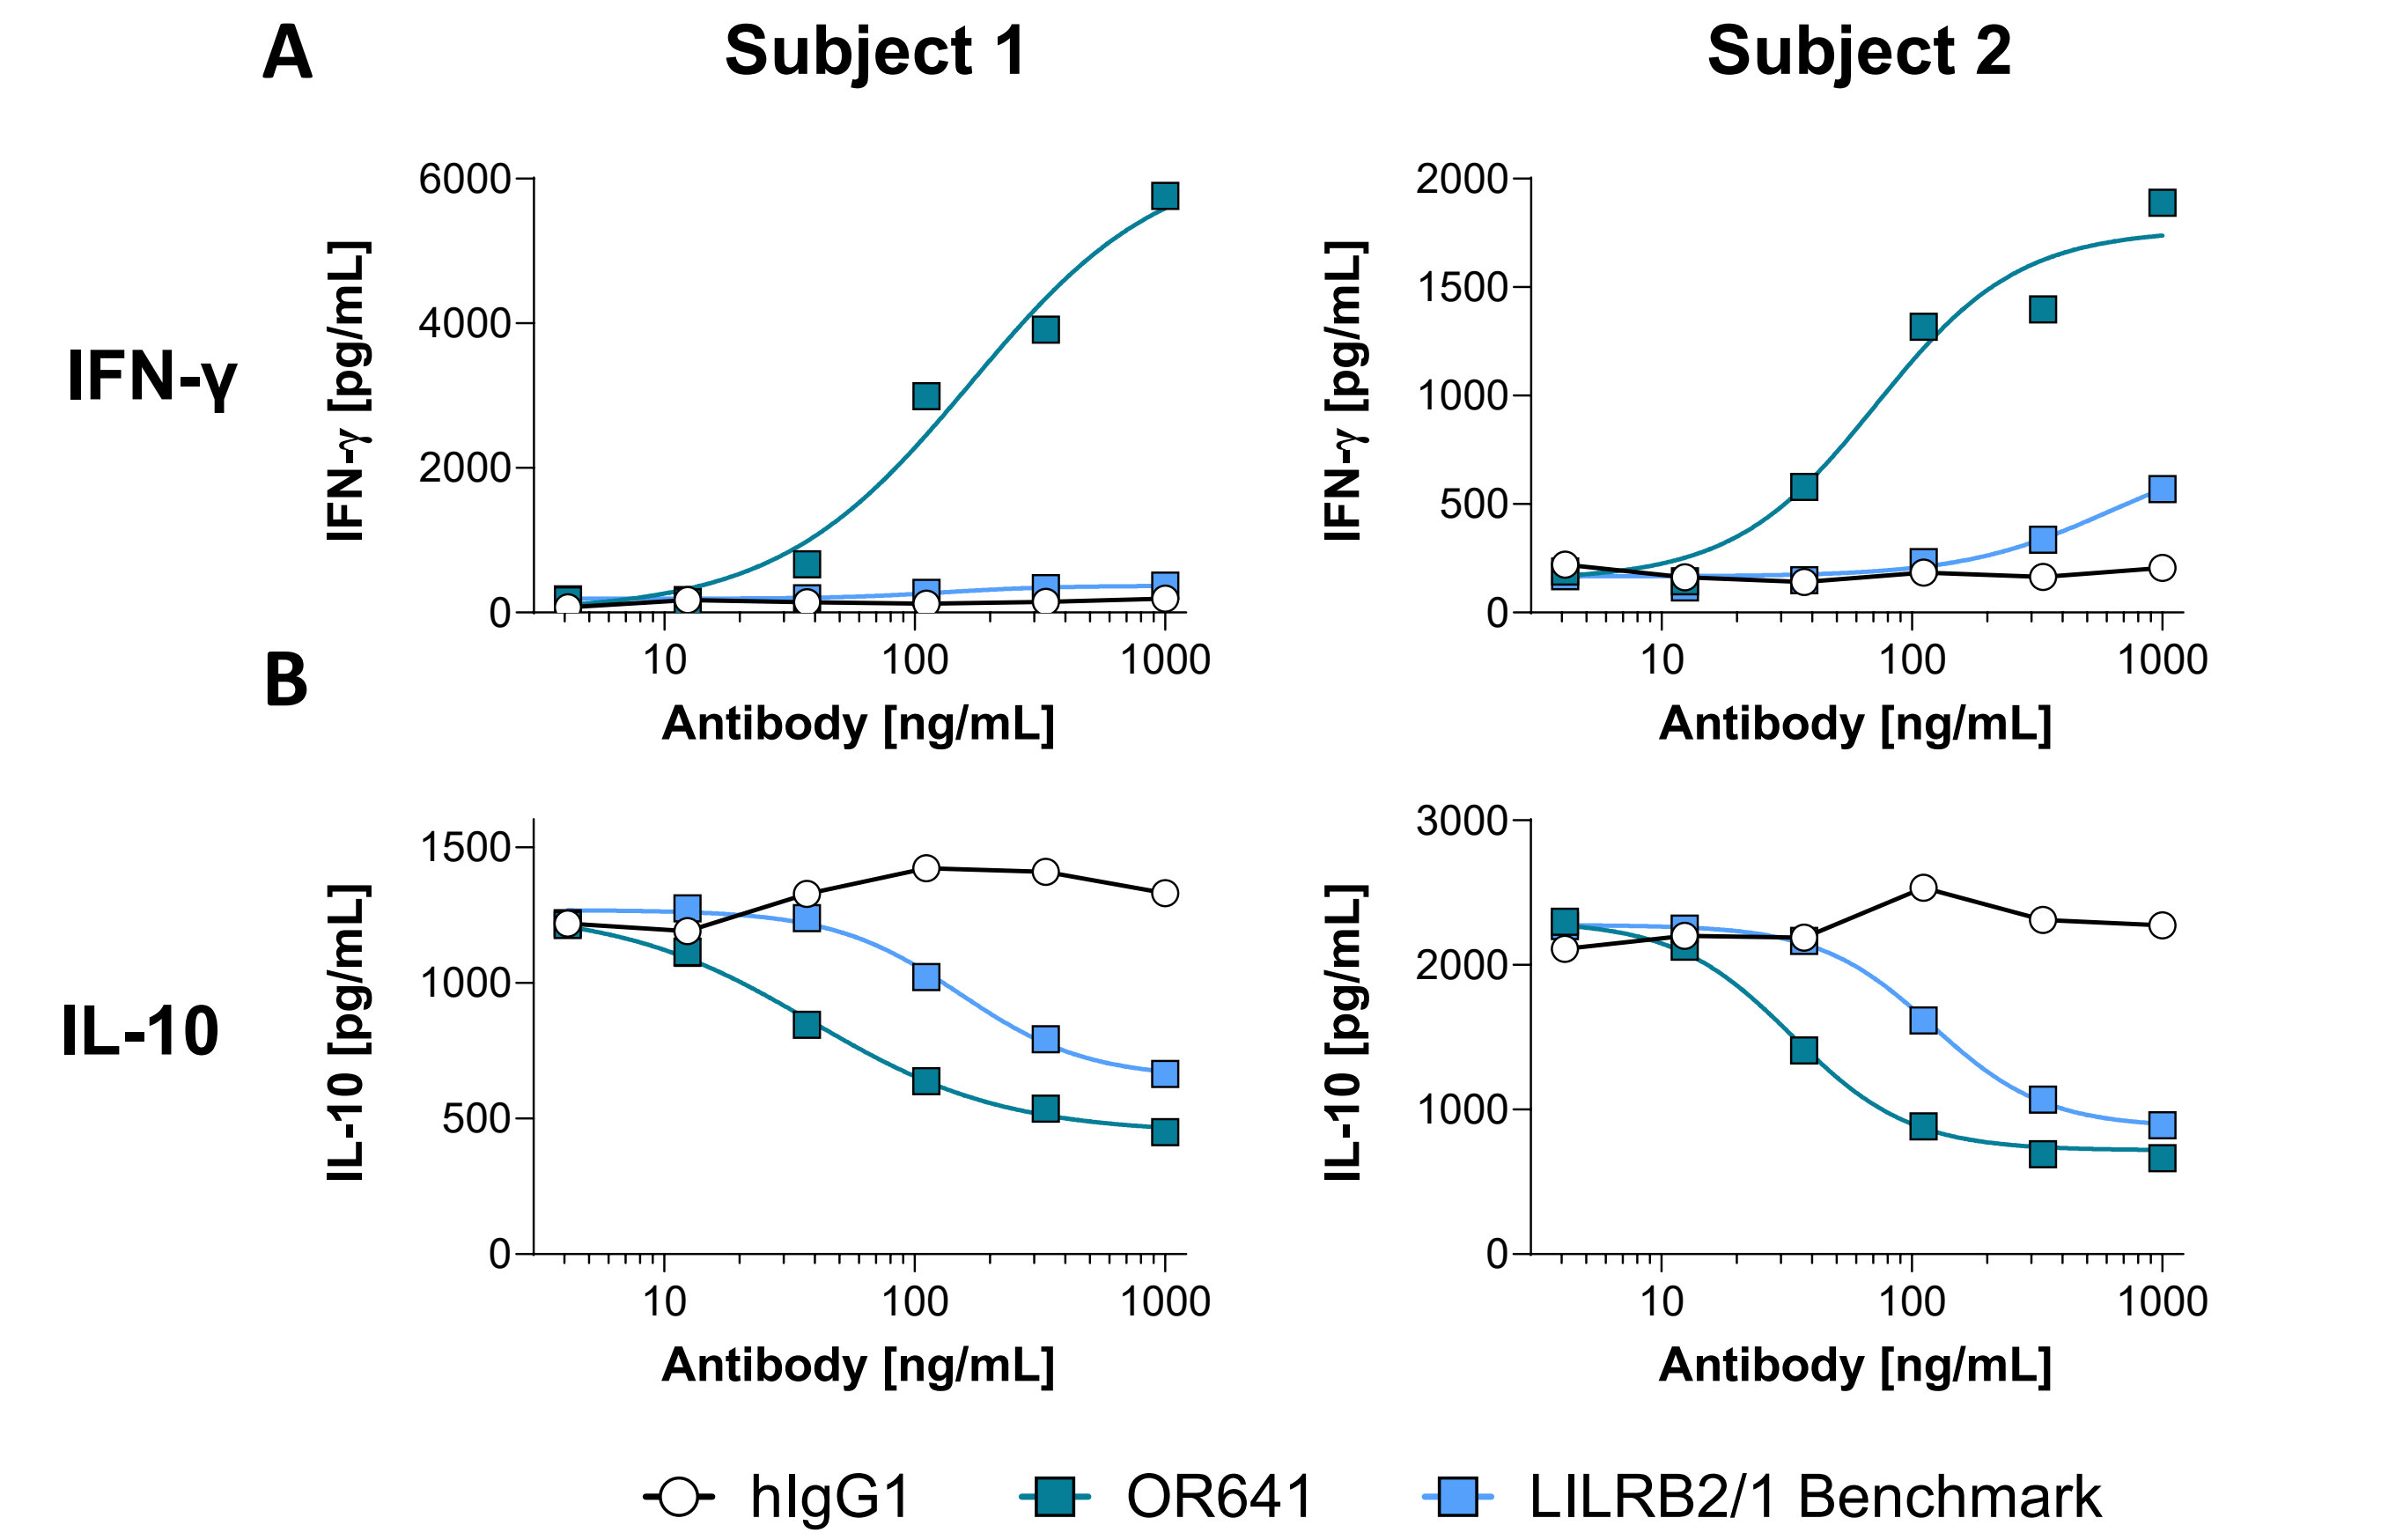
A
Subject 1
Subject 2
IFN-γ
IFN-γ [pg/mL]
B
IL-10
IL-10 [pg/mL]
Antibody [ng/mL]
hIgG1
OR641
LILRB2/1 Benchmark

Figure 5. Human PBMCs are treated with OR641, benchmark anti-LILRB2/1 antibody or human IgG1 isotype control, stimulated with LPS for 24 hours, then assayed for IFN-γ and IL-10 secretion. OR641 treatment enhances A) IFN-γ secretion and B) decreases IL-10 secretion in human PBMCs

OR641 enhances HLA-G mediated NK cell cytotoxicity

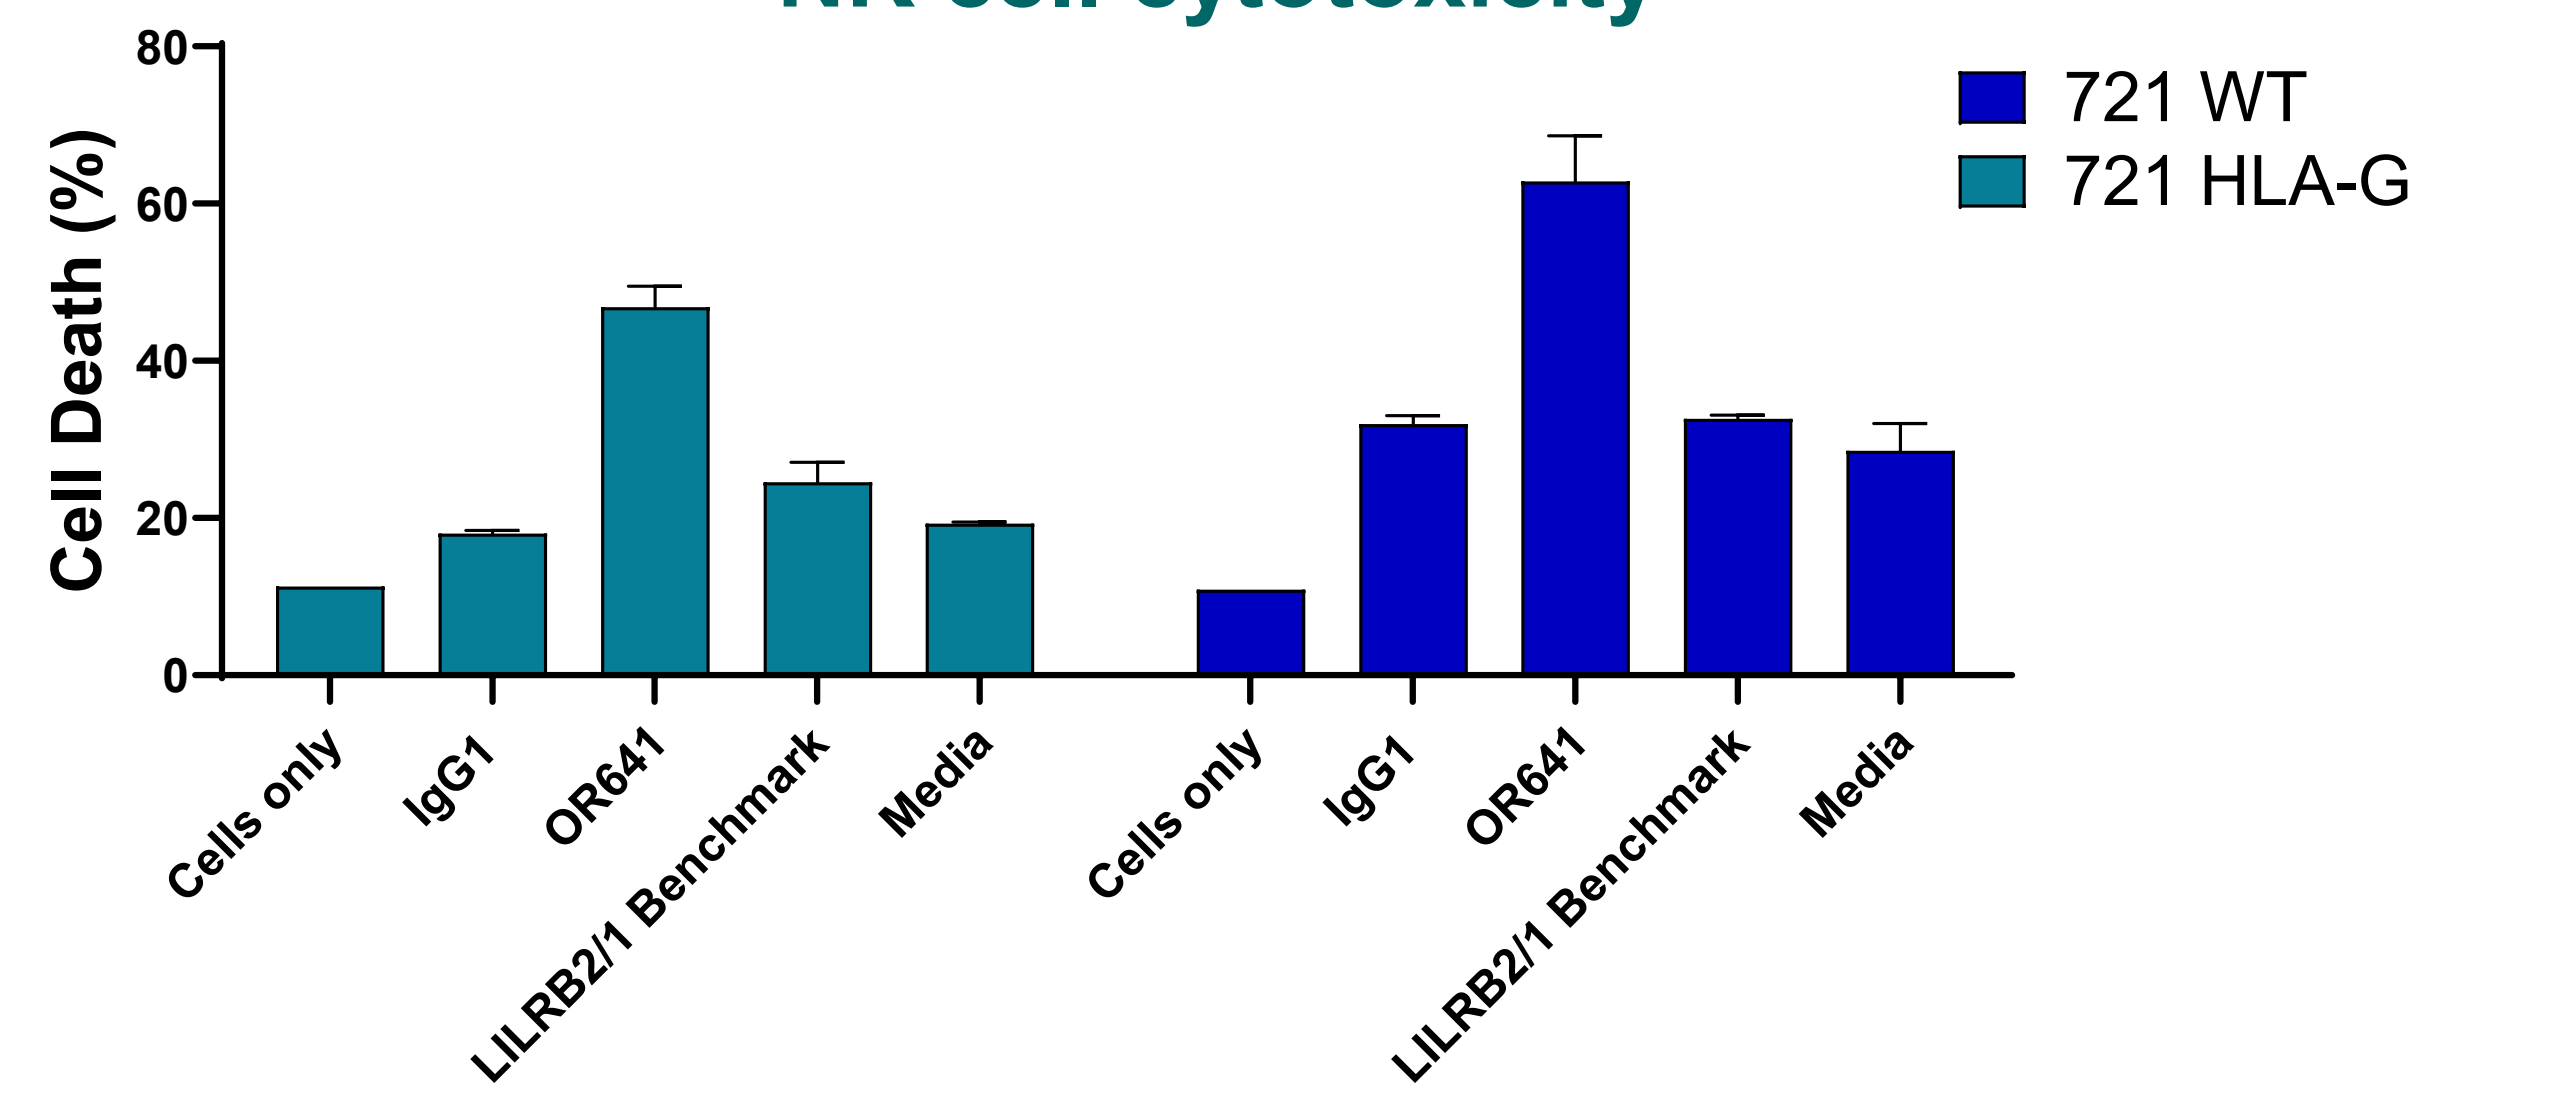
Cell Death (%)
721 WT
721 HLA-G
Cells only
IgG1
OR641
LILRB2/1 Benchmark
Media

Figure 6. Treatment with OR641 enhances NK cell mediated cytotoxicity of HLA-G expressing and wild type 721.221 B-cell lymphoma cells.

OR641 significantly enhances HLA-G mediated M2c macrophage phagocytosis

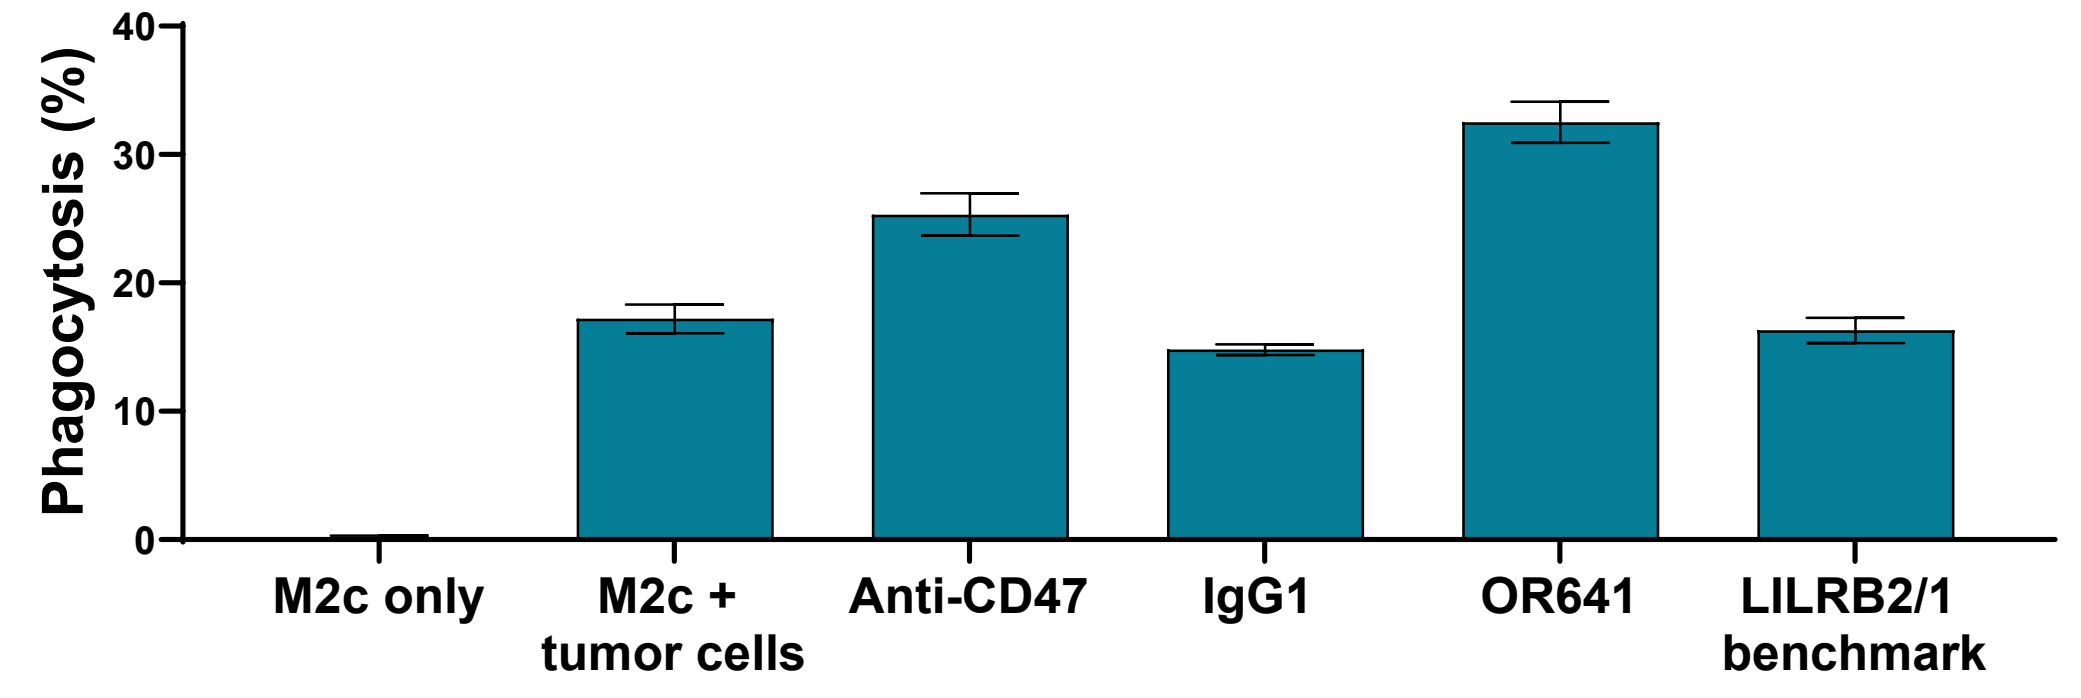
Phagocytosis (%)
M2c only
M2c + tumor cells
Anti-CD47
IgG1
OR641
LILRB2/1 benchmark

Figure 7. M2c macrophages were treated with OR641 or LILRB2/1 benchmark for 18 hours, then stained for phagocytosis of CellTace™ labeled 721.221 B lymphoma cells expressing HLA-G. OR641 treatment enhances HLA-G-mediated macrophage phagocytosis.

Summary

- OR641 binds to human LILRB1 and LILRB2, and blocks binding to HLA-G
- OR641 enhances IFN-γ secretion and decreases IL-10 secretion in PBMC stimulated with LPS
- OR641 modulates the immunosuppressive function of M2-like TAMs and enhances adaptive anti-tumor responses in M2c/T cell coculture assays
- OR641 enhances macrophage phagocytosis and NK cell mediated cytotoxicity of wild-type and HLA-G expressing lymphoma B-cells

Acknowledgements

We would like to acknowledge the contributions of Bob Lechleider, and Cliff Stocks at OncoResponse, our Scientific advisors: Michael Curran, David DeNardo, Mike Gallatin, David Hong, Miriam Merad, Anil Singhal, and James Welsh.

OncoResponse

http://www.oncoresponse.com

THE UNIVERSITY OF TEXAS MD Anderson Cancer Center